CHARACTERISTICS ASSOCIATED WITH BEING ADHERENT IN PATIENTS EXPERIENCING LATE REFILLS BEFORE AND AFTER IMPLEMENTATION OF A REFILL REMINDER PROGRAM

Authors: James Chamberlain, PharmD, MS[1]; Shellie Keast, PharmD, PhD[1]; Jennifer Hendricks, Pharm.D., APh, BCACP, CDCES, FCPhA[1]
Affiliations: 1. MedImpact Healthcare Systems, San Diego, CA, USA

INTRODUCTION

- Medication nonadherence is known to contribute to poor health outcomes and associated costs are estimated to exceed $100 billion in the US annually.[1,2]
- Pharmacy benefit managers (PBMs) are tasked with improving member engagement and promoting medication adherence.
- Refill reminder programs can increase members' likelihood of refilling their medications and maintaining their health by addressing the most common barrier reported by patients: forgetfulness.[2,3]
- Incremental improvements in medication adherence may also improve Medicare Star Ratings, which offer additional financial benefits to health plans.

OBJECTIVE

The objective of this study was to evaluate the association of various plan member characteristics and the outcome of having a proportion of days covered (PDC) greater than 80% (adherent) for three medication classes: diabetes (DM), hypertension (HTN), and statins in four Medicare Part D health plans with populations of less than 3,000 before and after the implementation of a refill reminder program.

METHODOLOGY:

- Two cohorts for calendar years 2023 (no refill reminder program) and 2024 (with refill reminder program) were identified.
- The inclusion criteria included:
 - Members were eligible in both 2023 and 2024
 - Filled a refill reminder target drug (once in the first and last half of the calendar year),
 - Have one late claim within a target drug group in the first six months of each year.
- Descriptive statistics and general linear regression for a binary outcome was used to model the odds of being adherent (PDC ≥ 80%) with member characteristics (year, age at study midpoint, sex, month of first late refill, drug class, use of multiple drug classes, use of 90-day supplies, total annual late refills, and total annual late refills refilled).

Figure 1: Attrition Flow Chart

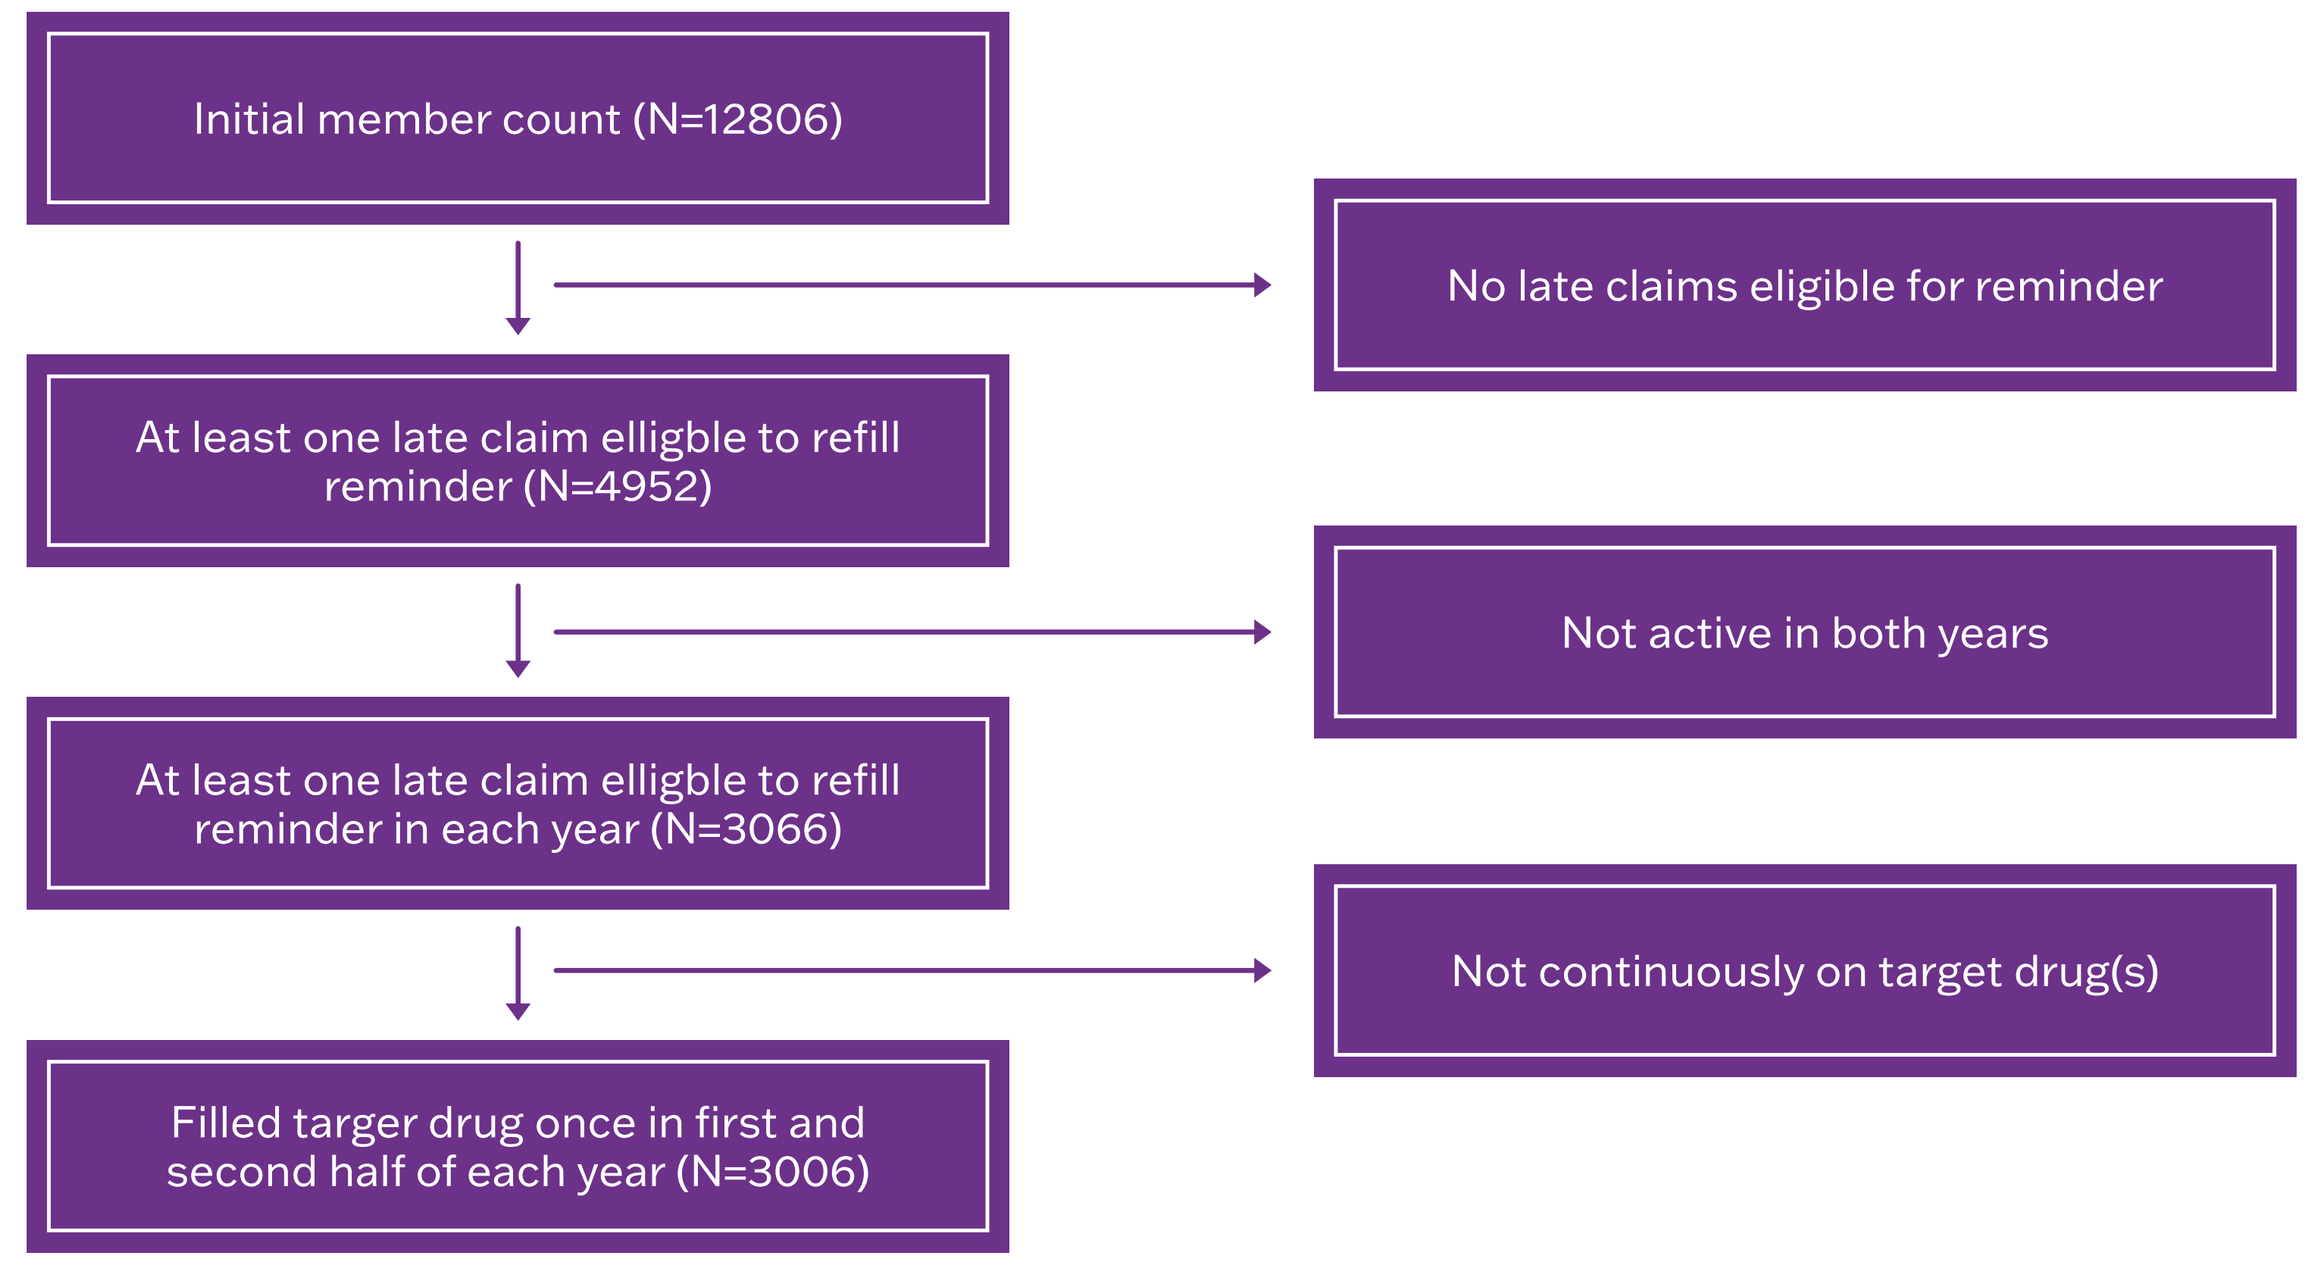
Initial member count (N=12806)
No late claims eligible for reminder
At least one late claim elligble to refill reminder (N=4952)
Not active in both years
At least one late claim elligble to refill reminder in each year (N=3066)
Not continuously on target drug(s)
Filled targer drug once in first and second half of each year (N=3006)

Figure 2: Study Flow Diagram

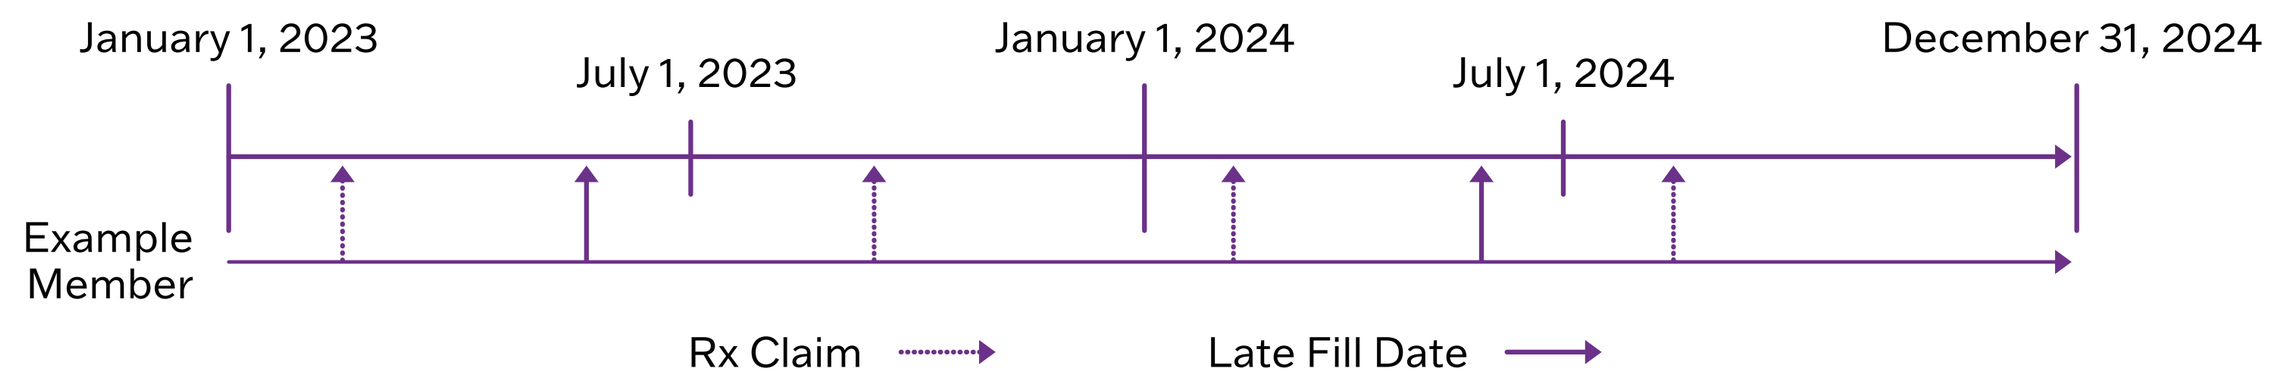
January 1, 2023
July 1, 2023
January 1, 2024
July 1, 2024
December 31, 2024
Example Member
Rx Claim
Late Fill Date

RESULTS

Table 1: Descriptive Characteristics by Year

Characteristic	2023	2024	P value
Age at Study Mid-Point			0.0483
Mean (SD)	75.40 (7.03)	75.9 (7.02)	
Sex, n (%)			0.7687
M	819 (51.48)	736 (52.01)	
F	772 (48.52)	679 (47.99)	
Month First Late Refill, n (%)			0.7396
February	138 (8.67)	129 (9.12)	
March	308 (19.36)	289 (20.42)	
April	443 (27.84)	364 (25.72)	
May	412 (25.90)	367 (25.94)	
June	290 (18.23)	266 (18.80)	
Use of 90 Day Fills			0.63
Yes	1,558 (97.93)	1,382 (97.67)	
No	33 (2.07)	33 (2.33)	
Medication Class,n (%)			0.0549
Diabetes	321 (20.18)	337 (23.82)	
Hypertension	626 (39.35)	531 (37.53)	
Statin	644 (40.48)	547 (38.66)	
Number of Classes, n (%)			0.1039
One	235 (14.77)	180 (12.72)	
Two or More	1,356 (85.23)	1,235 (87.28)	
Total Late Refills, mean (SD)	2.41 (1.68)	2.43 (1.57)	0.7584
Total Late Refilled, mean (SD)	0.70 (0.96)	0.67 (0.97)	0.4005
Adherent at ≥ 80%,n (%)			0.3103
Yes	1,358 (85.36)	1,226 (86.64)	
No	233 (14.64)	189 (13.36)	

Table 2: Results of Logistic Regression for Adherent vs. Non-Adherent

Characteristic 2023	OR (95% CI)	P value
Main		
2024 (vs 2023)	1.66 (0.94, 2.95)	0.0807
Age (Years, Continuous)	1.00 (0.98, 1.02)	0.7267
Sex (vs Male)	0.95 (0.75, 1.21)	0.6945
Month of First Late Refill (vs June)		
February	5.89 (2.02, 17.18)	0.0012
March	2.14 (1.21, 3.81)	0.0093
April	0.90 (0.57, 1.40)	0.633
May	1.15 (0.72, 1.82)	0.5567
Drug Group (vs Statin)		
Diabetes	2.46 (0.64, 9.50)	0.1911
Hypertension	1.94 (1.14, 3.31)	0.0144
More than One Drug Groups (vs Only One)	4.13 (2.82, 6.04)	<.0001
90 Day Use (vs 30 Day)	0.52 (0.13, 2.12)	0.3625
Number of Late Claims (Count)	0.29 (0.18, 0.47)	<.0001
Number of Late Claims Refilled (Count)	4.28 (3.10, 5.71)	<.0001
Interactions		
Interactions Year and Drug Group (vs 2023 and Statin)		
2024 and Diabetes	1.84 (1.02, 3.31)	0.0437
2024 and Hypertension	0.87 (0.55, 1.37)	0.5387
Sex and Drug Group (vs Male and Statin)		
Female and Diabetes	0.51 (0.27, 0.95)	0.0337
Female and Hypertension	0.84 (0.52, 1.34)	0.4624
Drug Group and Number of Drug Groups (vs Statin and Only One)		
Diabetes and More than One Drug Group	0.23 (0.06,0.87)	0.0305
Hypertension and More than One Drug Group	0.72 (0.39, 1.30)	0.2743
Number of Late Claims and Drug Group (vs Statin)		
Number of Late Claims and Diabetes	1.36 (1.11, 1.68)	0.0032
Number of Late Claims and Hypertension	1.03 (0.90, 1.18)	0.6951
Number of Late Claims and 90 Day Use (vs 30 Day)	2.22 (1.38, 3.56)	0.0009
Number of Late Claims and Number of Late Claims Refilled	0.9 (0.85, 0.95)	<.0001

Figure 3. Percent of Members Adherent at > 80% by Year

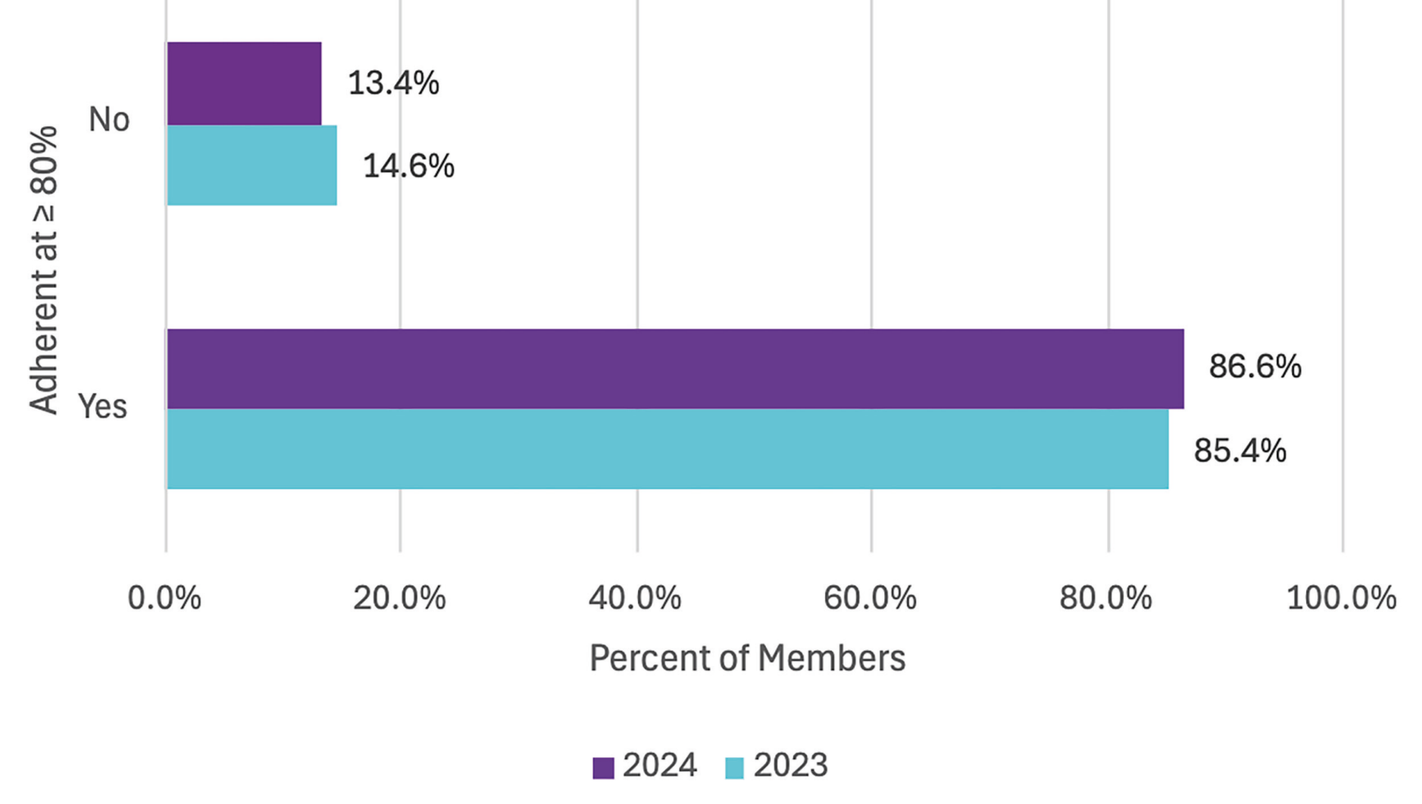
Adherent at ≥ 80%
No
13.4%
14.6%
Yes
86.6%
85.4%
0.0% 20.0% 40.0% 60.0% 80.0% 100.0%
Percent of Members
2024 2023

Figure 4. Percent of Members on Target Drug Classes by Year

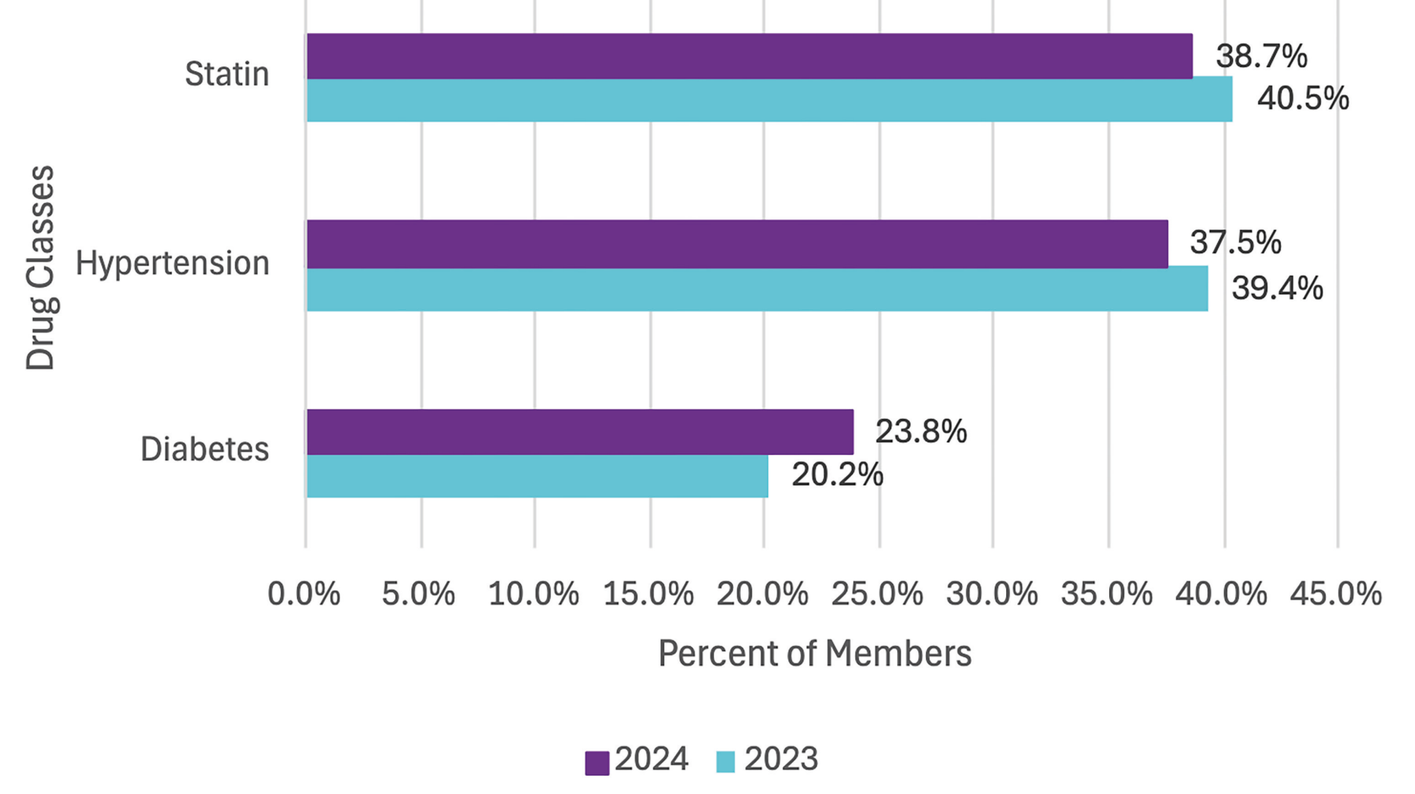
Drug Classes
Statin
38.7%
40.5%
Hypertension
37.5%
39.4%
Diabetes
23.8%
20.2%
0.0% 5.0% 10.0% 15.0% 20.0% 25.0% 30.0% 35.0% 40.0% 45.0%
Percent of Members
2024 2023

Figure 5. Percent of Members on Multiple Drug Classes by Year

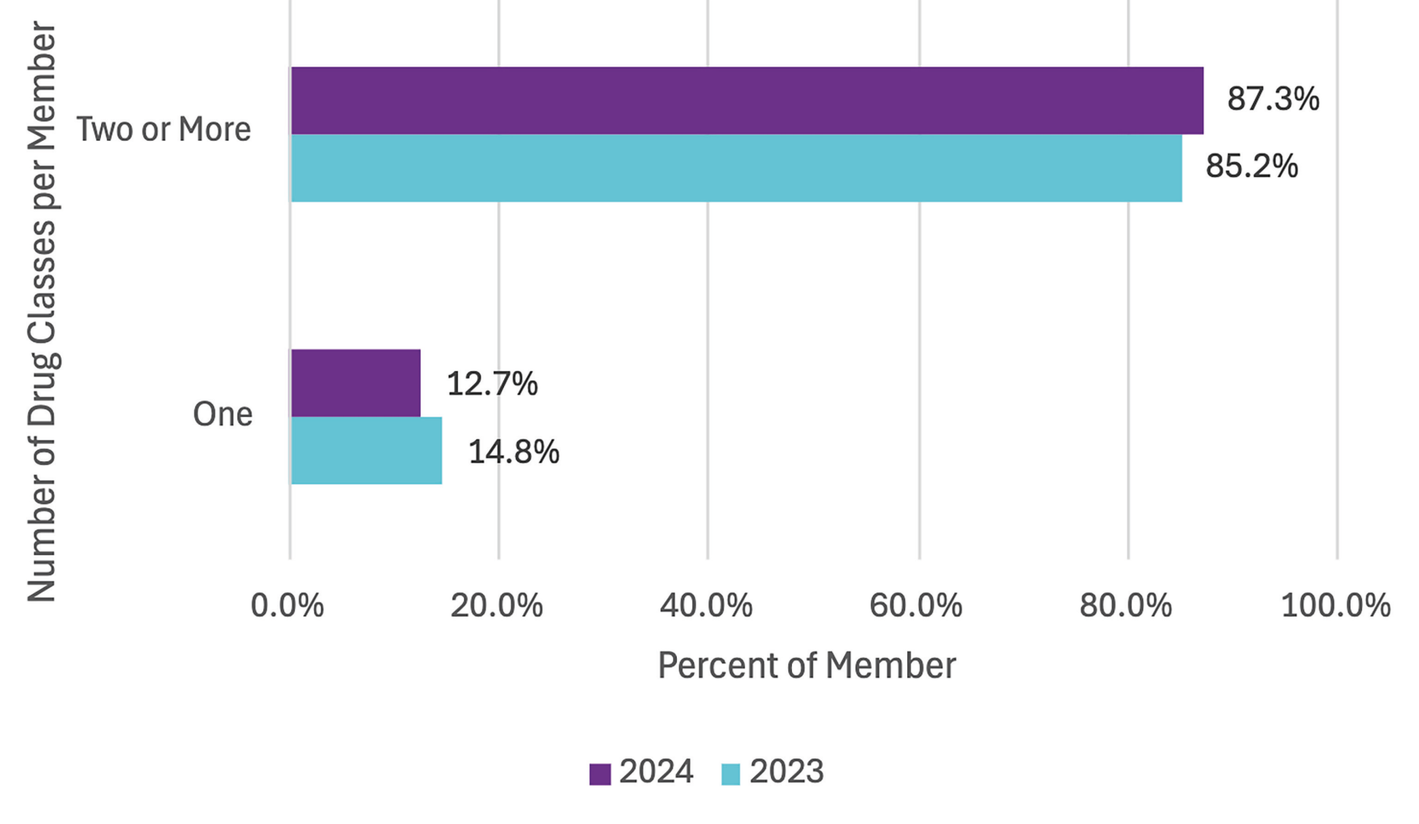
Number of Drug Classes per Member
Two or More
87.3%
85.2%
One
12.7%
14.8%
0.0% 20.0% 40.0% 60.0% 80.0% 100.0%
Percent of Member
2024 2023

Figure 6.Figure 6. Odds Ratios and 95% CIs for Select Factors

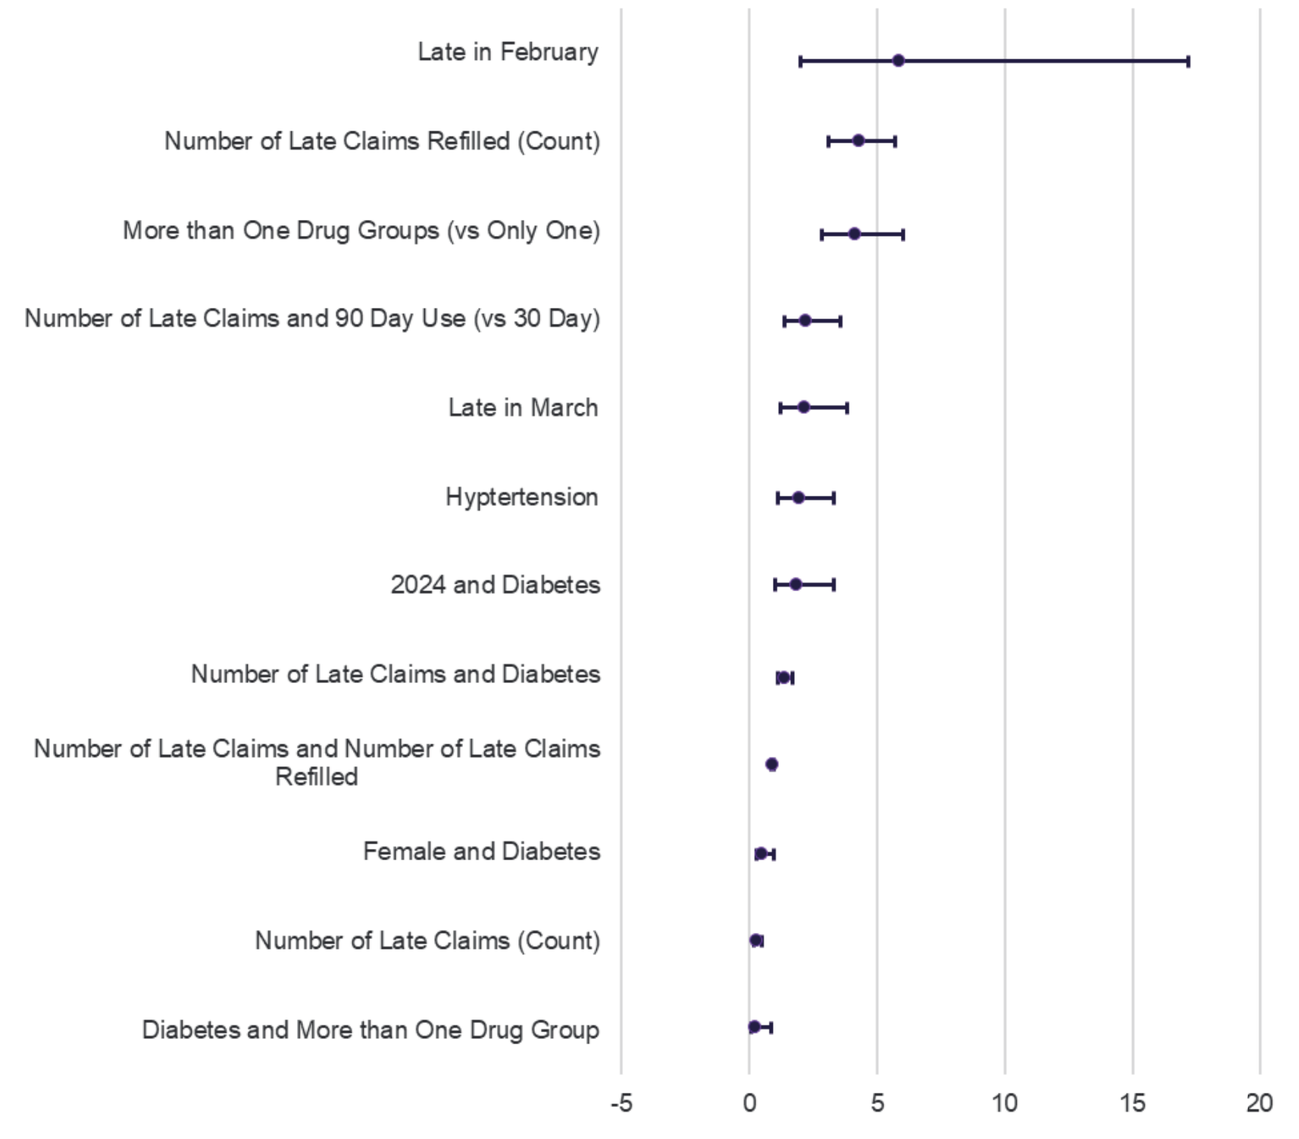
Late in February
Number of Late Claims Refilled (Count)
More than One Drug Groups (vs Only One)
Number of Late Claims and 90 Day Use (vs 30 Day)
Late in March
Hyptertension
2024 and Diabetes
Number of Late Claims and Diabetes
Number of Late Claims and Number of Late Claims Refilled
Female and Diabetes
Number of Late Claims (Count)
Diabetes and More than One Drug Group
-5 0 5 10 15 20

DISCUSSION

- Overall odds of being adherent were not significant for year 2024 compared to 2023, however those who had a late refill in February or March had increased odds of being adherent, which might be related to early intervention at a pharmacy or via the refill reminder program. It might also be a result of having more time in the year to make up for a late refill.
- Compared to statins, members on hypertension medications had an increased odds (1.9) of being adherent. Additionally, members having more than one drug group had an odds of being adherent 4.1 times that of those only having one drug group, which could be due to multiple opportunities for intervention or member concern for overall more complicated health.
- Having more late claims overall decreased the odds of being adherent (0.29), while refilling those late claims increased the odds of being adherent by 4.3 times. This indicates the importance of intervention for patients who are experiencing delayed refills.
- Looking at some interactions related to diabetes medications, we see that being in year 2024 and having diabetes medications had an increased odds (1.8) of being adherent. This could be due to the introduction of the refill reminder program or possibly the increased use of GLP-1 medications. Interestingly, being female and on diabetes medications had a decreased odds of being adherent (0.51) this might also be due to the increased use of GLP-1s and a higher discontinuation rate for these medications. Of note is that being on a diabetes medication and in multiple drug groups had a lower odds of being adherent (0.23) compared to being on a statin alone. And finally, we see that increasing number of late claims and being on diabetes medication had an increased odds of being adherent (1.4) compared to statins again possibly due to an increased opportunity for interventions and members considering diabetes as a more serious disease.
- Two final interactions of note are for number of late claims and 90-day use or number of refilled claims. Those who had increased late claims and used 90-day supplies had an increased odds of being adherent (2.2) compared to those on 30-day supplies indicating that the use of 90-day supplies is a beneficial tool in improving adherence. For each additional late claim and late claim refilled, the odds were decreased by a factor of 0.9, which demonstrates that an increasing late claim count will eventually lower adherence even when the claims are subsequently refilled.
- Limitations
 - Results of the analysis are from a single population and may not be generalizable to the larger population.
 - This study used administrative claims data collected for the primary purpose of billing and reimbursement of prescriptions and may contain errors.

CONCLUSION

Adherence involves complicated factors related to the individual's unique characteristics. In this study refilling late medications, being on multiple drug groups, and using 90-day supplies were associated with increased odds of being adherent. Being in the 2024 cohort and on a DM medication also had increased odds of adherence. Further research is needed to explore the effects of the refill reminder program on adherence in similar populations.

REFERENCES

1. Peasah, S. K., Liu, Y., Krohe, S., Campbell, V., Lee, C., Mathur, A., Stevenson, H., Manolis, C., & Good, C. B. (2024). Assessing the impact of a financial incentive and refill reminder program on medication adherence and costs. Journal of managed care & specialty pharmacy, 30(1), 43–51. https://doi.org/10.18553/jmcp.2024.30.1.43
2. Neiman, A. B., Ruppar, T., Ho, M., Garber, L., Weidle, P. J., Hong, Y., George, M. G., & Thorpe, P. G. (2017). CDC Grand Rounds: Improving Medication Adherence for Chronic Disease Management - Innovations and Opportunities. MMWR. Morbidity and mortality weekly report, 66(45), 1248–1251. https://doi.org/10.15585/mmwr.mm6645a2
3. Taitel, M. S., Mu, Y., Gooptu, A., & Lou, Y. (2017). Impact of late-to-refill reminder calls on medication adherence in the Medicare Part D population: evaluation of a randomized controlled study. Patient preference and adherence, 11, 373–379. https://doi.org/10.2147/PPA.S127997